The Effects Of AQX-1125, A Selective Oral SHIP1 Activator, On Lipopolysaccharide-Induced Cellular And Biochemical Changes In Sputum From Healthy Volunteers

J. S. Elborn, MD[1], S.P. Smith, MD[2], V. Brown, MD[1], D. Chernoff, MD[3], P. Tam, PhD[3], J. Neville , PhD[3], J. Toews, MSc[3], G.R. Stenton , PhD[3], C. Harwig , PhD[3], T. MacRury , PhD[3], and L.F. Mackenzie, MSc[3]

[1]Queen's University Belfast, UK; [2]Celerion, Belfast, UK; [3]Aquinox Pharmaceuticals Inc., Richmond, Canada

A1509

Abstract

Rationale: SH2-containing inositol-5'-phosphatase 1 (SHIP1) down-regulates the pro-inflammatory phosphoinositide 3-kinase (PI3K)/Akt pathway by metabolizing PI(3,4,5)P3 to PI(3,4)P2. SHIP1 activation is an established pharmacological approach for the control of inflammatory disorders.

Objective: To determine the effects of SHIP1 activation by AQX-1125 on lipopolysaccharide (LPS)-induced lung inflammation in healthy volunteers.

Methods: In this single-center, placebo-controlled, two-way crossover study, 18 healthy volunteers were randomised to one of two treatment sequences (placebo then AQX-1125 or AQX-1125 then placebo) in a double-blind fashion. In each treatment period subjects received either oral AQX-1125 (450 mg) or matching placebo once-daily, for 7 days. The wash-out period was 28 days between the two treatment periods. The primary endpoint was the effect of AQX-1125 on sputum neutrophilia 24 h after aerosol LPS challenge. Secondary endpoints included sputum biomarker and PK analysis, as well as safety and tolerability assessment.

Results: AQX-1125 significantly reduced sputum neutrophil counts 24 h following the inhaled LPS-challenge when compared to placebo by ANOVA (p=0.014). The absolute sputum cell counts in subjects treated with AQX-1125 (1.4 x 10^6 cells/gram sputum) were reduced by 67% when compared to sputum cell counts from subjects treated with placebo (4.3 x 10^6 cells/gram sputum). When baseline sputum neutrophils were included as a covariate in the analysis (ANCOVA), AQX-1125 reduced the 24 h post-LPS sputum neutrophils by 62% (p=0.0615). The reduction of neutrophils correlated with a 62% reduction in the pre-dose Day 7 sputum IL-6 concentration (p=0.048). AQX-1125 was safe and well tolerated. The most frequent adverse events were related to the inhaled LPS challenge. The time to maximum plasma concentration at steady state ($C_{max,ss}$) indicated that AQX-1125 was rapidly absorbed. The mean $C_{max,ss}$ and AUC_{0-24} values were 1350 ng/mL and 16900 h.ng/mL respectively.

Conclusion: AQX-1125 has anti-inflammatory effects in healthy volunteers undergoing LPS-induced lung inflammation; reducing the LPS-mediated increases in sputum neutrophilia and suppressing sputum IL-6 concentrations. This clinical data demonstrates the efficacy of AQX-1125 in a standard pulmonary inflammation model and coupled with the safety, tolerability and PK data support the continued clinical development of the SHIP1 activator AQX-1125 for inflammatory disease.

Introduction

Targeting SHIP1

- PI3K pathway is an established target for drug development
- PI3K/SHIP1 pathway plays a key role in regulating cell migration and activation
- Targeting SHIP1 is an alternate way of modulating the PI3K pathway
- SHIP1 expression restricted to hematopoietic derived cells - limits off-target toxicity
- SHIP1 activation redirects cellular PI3K signaling, rather than preventing it

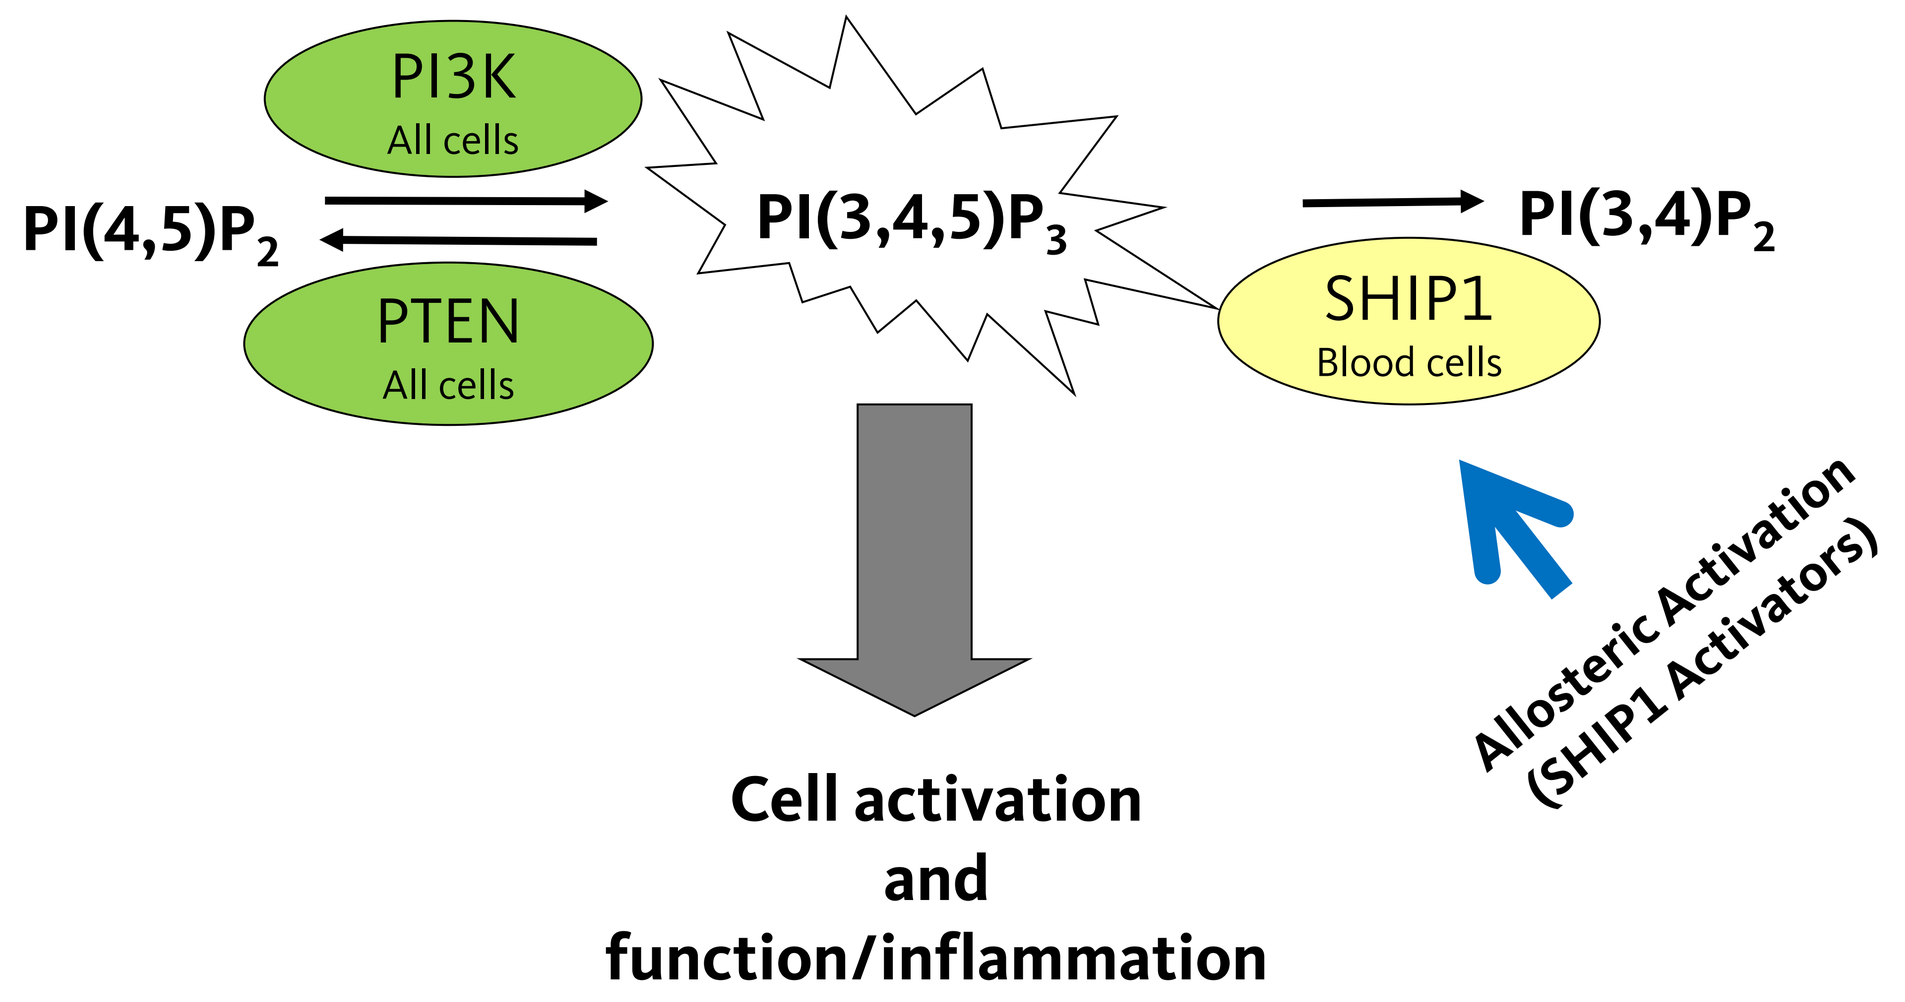
PI3K
All cells
PI(4,5)P2
PTEN
All cells
PI(3,4,5)P3
PI(3,4)P2
SHIP1
Blood cells
Allosteric Activation
(SHIP1 Activators)
Cell activation
and
function/inflammation

Figure 1. SHIP1 and PI3K signalling. SHIP1 activators redirect PI3K signalling, PI3K inhibitors block PI3K signalling.

Background

SHIP1 Activation

AQX-1125 is a small molecule next generation SHIP1 activator. It has many of the biological effects of the earlier generation SHIP1 activators[1,2], but has an improved drug scaffold and superior drug-like properties. These small molecules activate SHIP1 through an allosteric mechanism, via interaction with the C2 domain, and are anti-inflammatory in cellular and murine models.

An initial Phase 1 trial was conducted in healthy human subjects to study the safety, tolerability and pharmacokinetics of single ascending doses (17-542 mg), multiple ascending doses (100-542 mg for 10 days) and food effects (200 mg) of AQX-1125. The results showed that AQX-1125 was well tolerated at all doses, exhibited predictable pharmacokinetic behavior and absorbed equally in a fed versus a fasted state.

Study Design

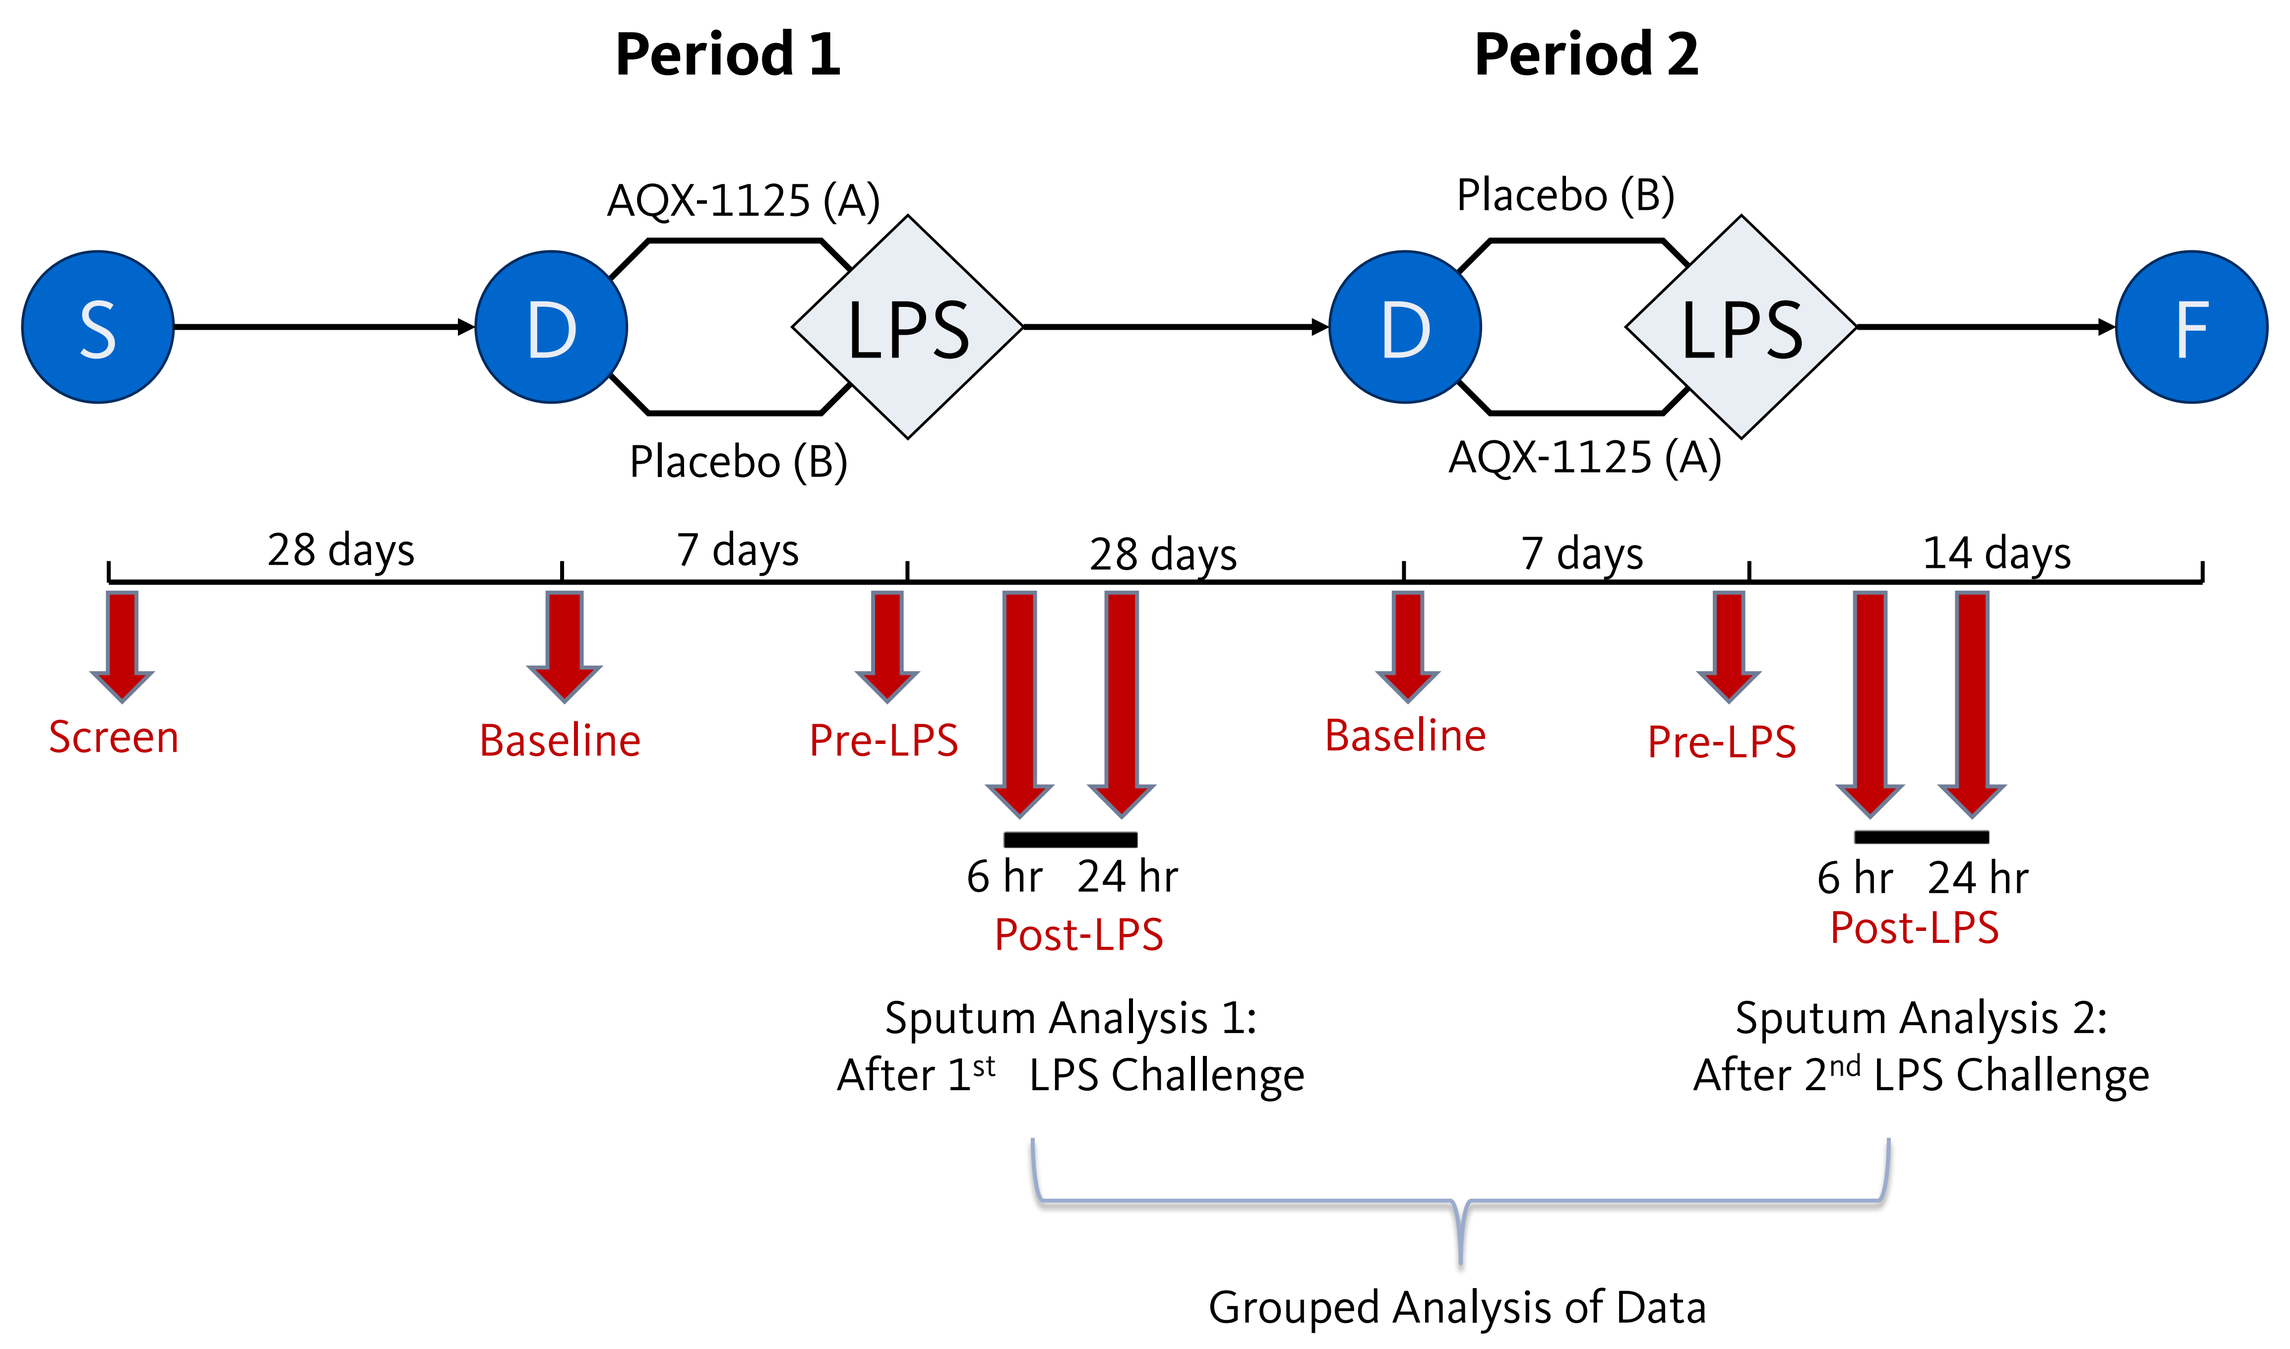
Period 1
Period 2
AQX-1125 (A)
Placebo (B)
S
D
LPS
F
Placebo (B)
AQX-1125 (A)
28 days
7 days
28 days
7 days
14 days
Screen
Baseline
Pre-LPS
6 hr 24 hr
Post-LPS
Baseline
Pre-LPS
6 hr 24 hr
Post-LPS
Sputum Analysis 1:
After 1st LPS Challenge
Sputum Analysis 2:
After 2nd LPS Challenge
Grouped Analysis of Data

Figure 2. The effects of AQX-1125 on LPS-induced cellular and biochemical changes in sputum were evaluated using a 2-way crossover design with a 28 day washout period between treatment periods. In each treatment period either 450 mg AQX-1125 or matching placebo was administered once daily for 7 days.

AQX-1125 was evaluated using LPS challenge in healthy human subjects in a two-way crossover design

Results

Characteristics	Mean (min-max)
Age (yr)	30 (21-44)
Height (m)	175 (158-190)
Body Weight (kg)	79 (60-103)
Mean BMI (kg/m²)	26 (21-30)

Table 1. A total of 18 healthy human subjects (15 males and 3 females) with a mean age of 30 years were randomized into this study. The mean (range) height, weight and body mass index (BMI) are shown. All subjects completed the study with no withdrawals.

Efficacy Analysis: Kinetics of Sputum Neutrophil induction

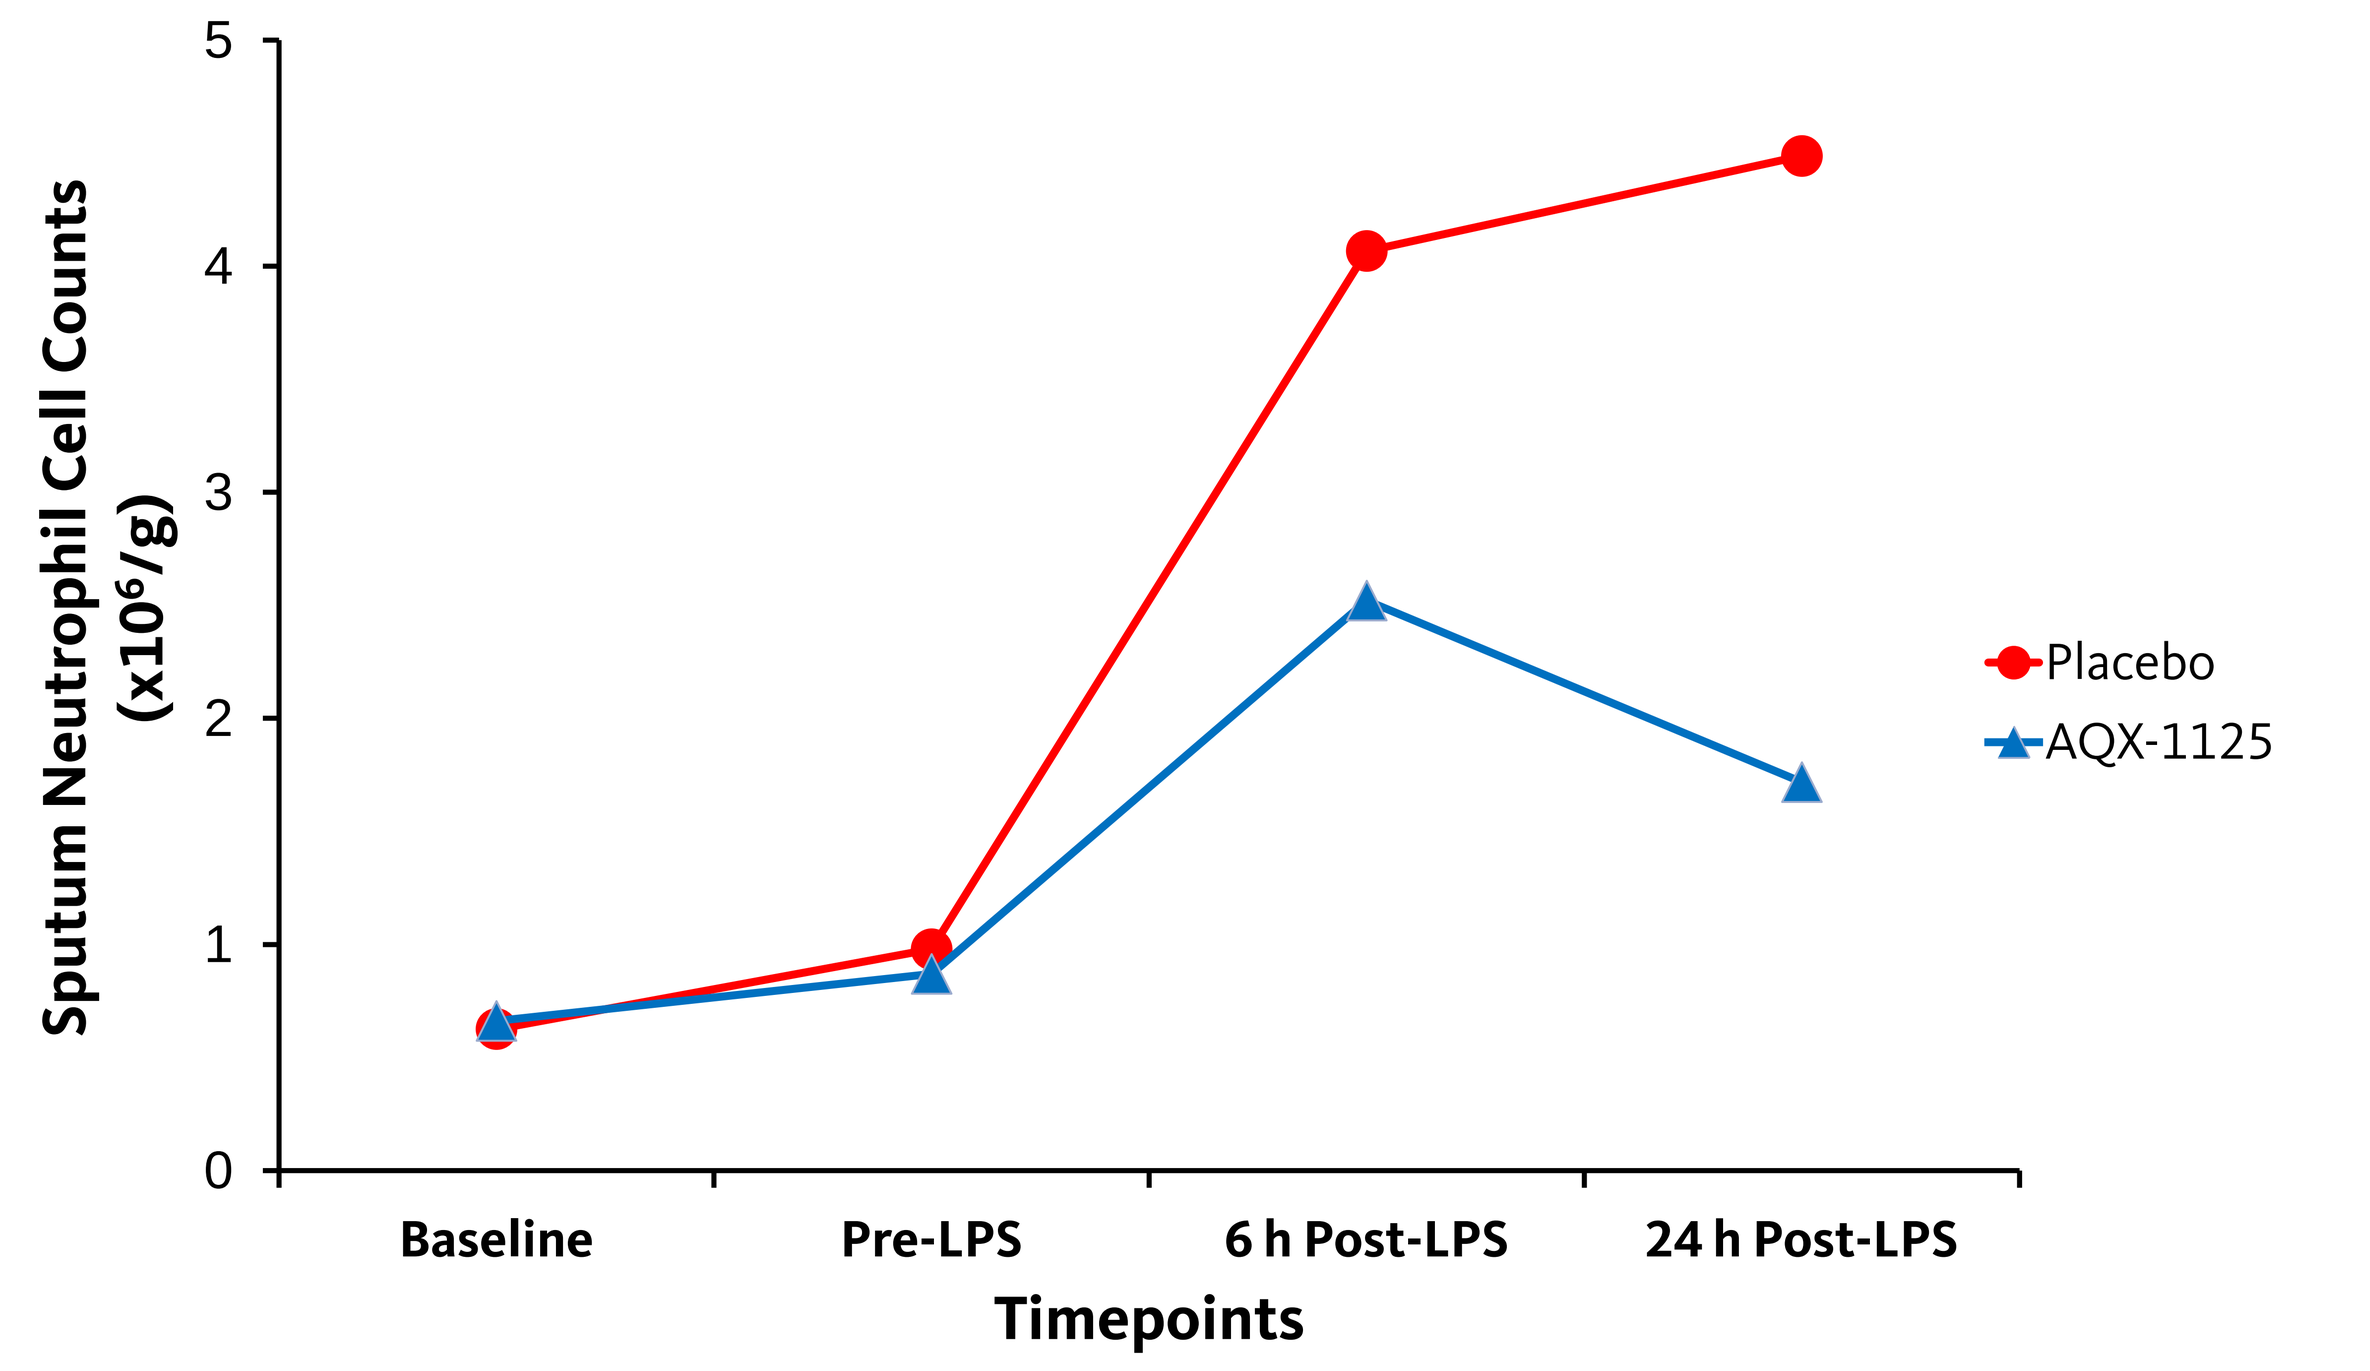
Sputum Neutrophil Cell Counts
(x10^6/g)
5
4
3
2
1
0
Baseline
Pre-LPS
6 h Post-LPS
24 h Post-LPS
Timepoints
Placebo
AQX-1125

Figure 3. The effect of AQX-1125 on the kinetics of LPS-induced neutrophilia in induced sputum. Mean sputum neutrophil counts in subjects on active and placebo treatment demonstrated a similar profile at baseline and just prior to inhaled LPS challenge. Following the challenge, a clear induction of sputum neutrophils occurred starting at 6 h post-LPS challenge. Mean sputum neutrophil counts were further increased in subjects on placebo treatment at 24 h. In comparison, subjects on active treatment demonstrated a decline in mean sputum neutrophils at 24 h when compared to the 6 h post-LPS timepoint. The statistical comparison between treatments is presented in *Table 2*.

AQX-1125 inhibited the accumulation of sputum neutrophils following inhaled LPS-challenge

Efficacy Analysis: Primary Endpoint – Sputum Neutrophils

Analysis	Timepoint	Sputum Neutrophil Counts AQX-1125	Sputum Neutrophil Counts Placebo	Treatment Ratio (Active/Placebo)	P-value
ANOVA	Baseline	0.440	0.638	0.69	0.491
	Pre-LPS	0.783	0.748	1.05	0.931
	6 h Post-LPS	2.269	4.086	0.56	0.226
	24 h Post-LPS	1.403	4.269	0.33	0.014
ANCOVA	Pre-LPS	0.667	0.714	0.93	0.914
	6 h Post-LPS	3.343	3.878	0.86	0.771
	24 h Post-LPS	1.606	4.271	0.38	0.062

Table 2. AQX-1125 significantly reduced the LPS-induced pulmonary inflammation as measured by the level of sputum neutrophils at the 24 h post-LPS challenge timepoint. There were no difference between treatment groups detected at baseline or at pre-LPS challenge. The comparisons between treatments were carried out on log transformed cell counts using the analysis of variance (ANOVA) and analysis of covariance (ANCOVA) with baseline used as the covariate. The back converted least-square mean for each timepoint for AQX-1125 and placebo treatment are shown with the treatment effect expressed as the ratio of the back converted least-square mean.

The primary objective was met with a 67% reduction (p=0.014) in sputum neutrophils at 24 hours induced by inhaled LPS challenge

Efficacy Analysis: Sputum IL-6 Concentrations

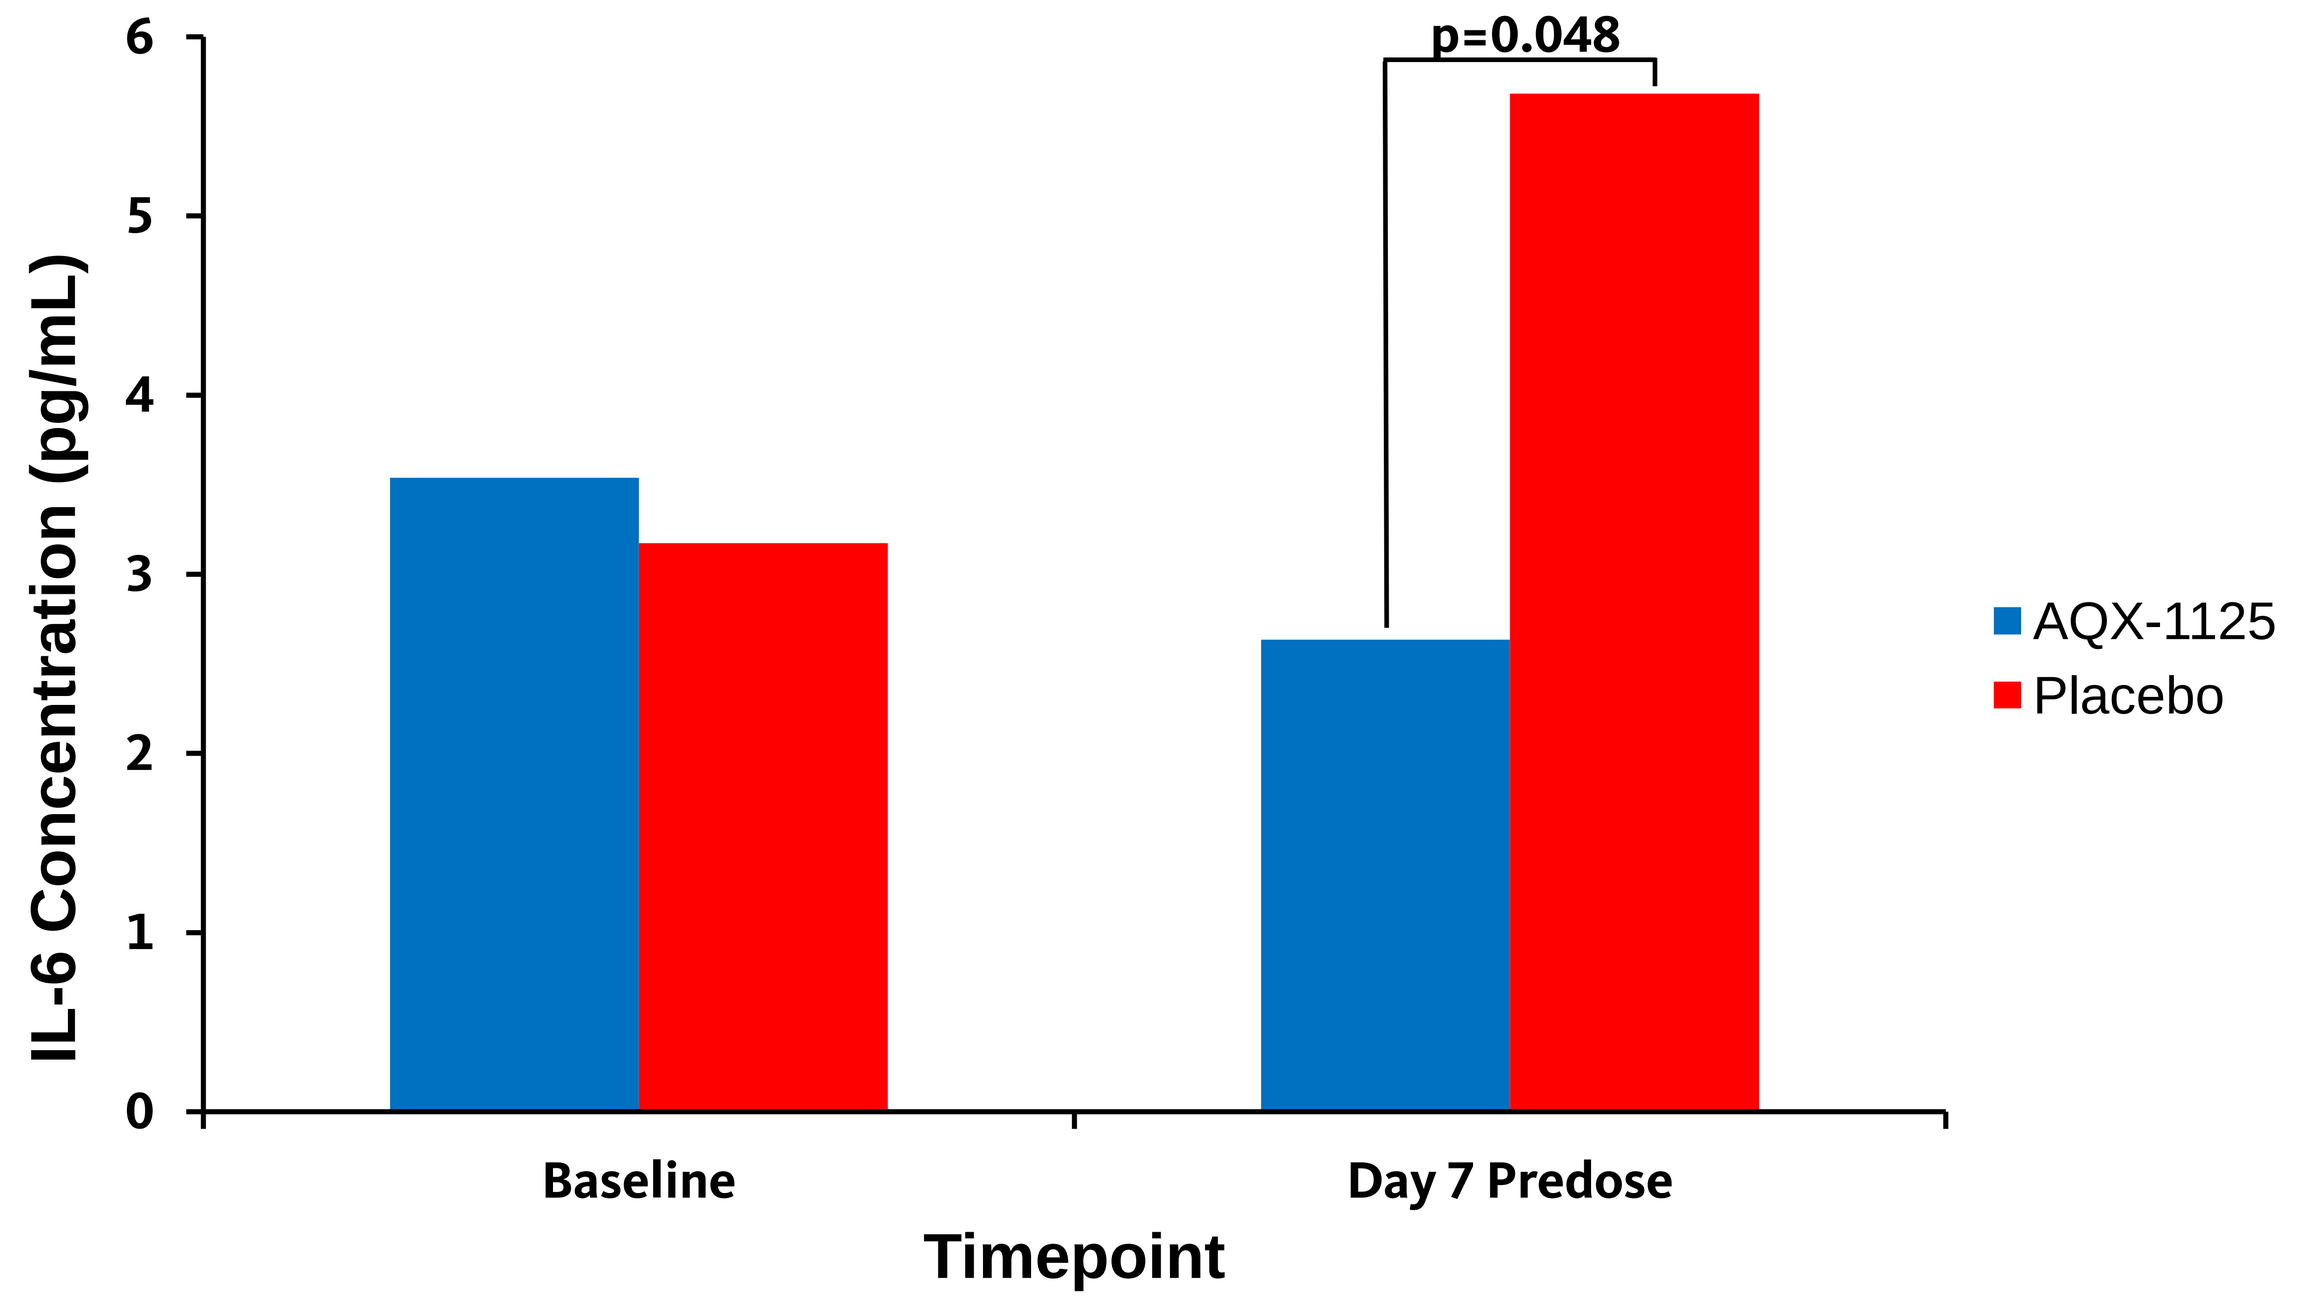
IL-6 Concentration (pg/mL)
6
5
4
3
2
1
0
p=0.048
Baseline
Day 7 Predose
Timepoint
AQX-1125
Placebo

Figure 4. The effect of AQX-1125 treatment on IL-6 concentrations in sputum. The effect of AQX-1125 on the primary endpoint was correlated with an AQX-1125 treatment effect on sputum IL-6 concentrations. On Day 7, the mean sputum IL-6 concentration following AQX-1125 treatment was reduced by 62% (p=0.048) when compared to placebo. No treatment difference in mean sputum IL-6 concentration was observed at baseline. No treatment differences were observed for sputum myeloperoxidase and IL-8 concentrations. Soluble analytes in sputum samples were analyzed using enzyme linked immunosorbent assay method. The comparisons between treatments were analyzed by ANOVA/ANCOVA as described above.

AQX-1125 mediated a 62% reduction (p=0.048) in sputum IL-6 concentrations

Pharmacokinetics

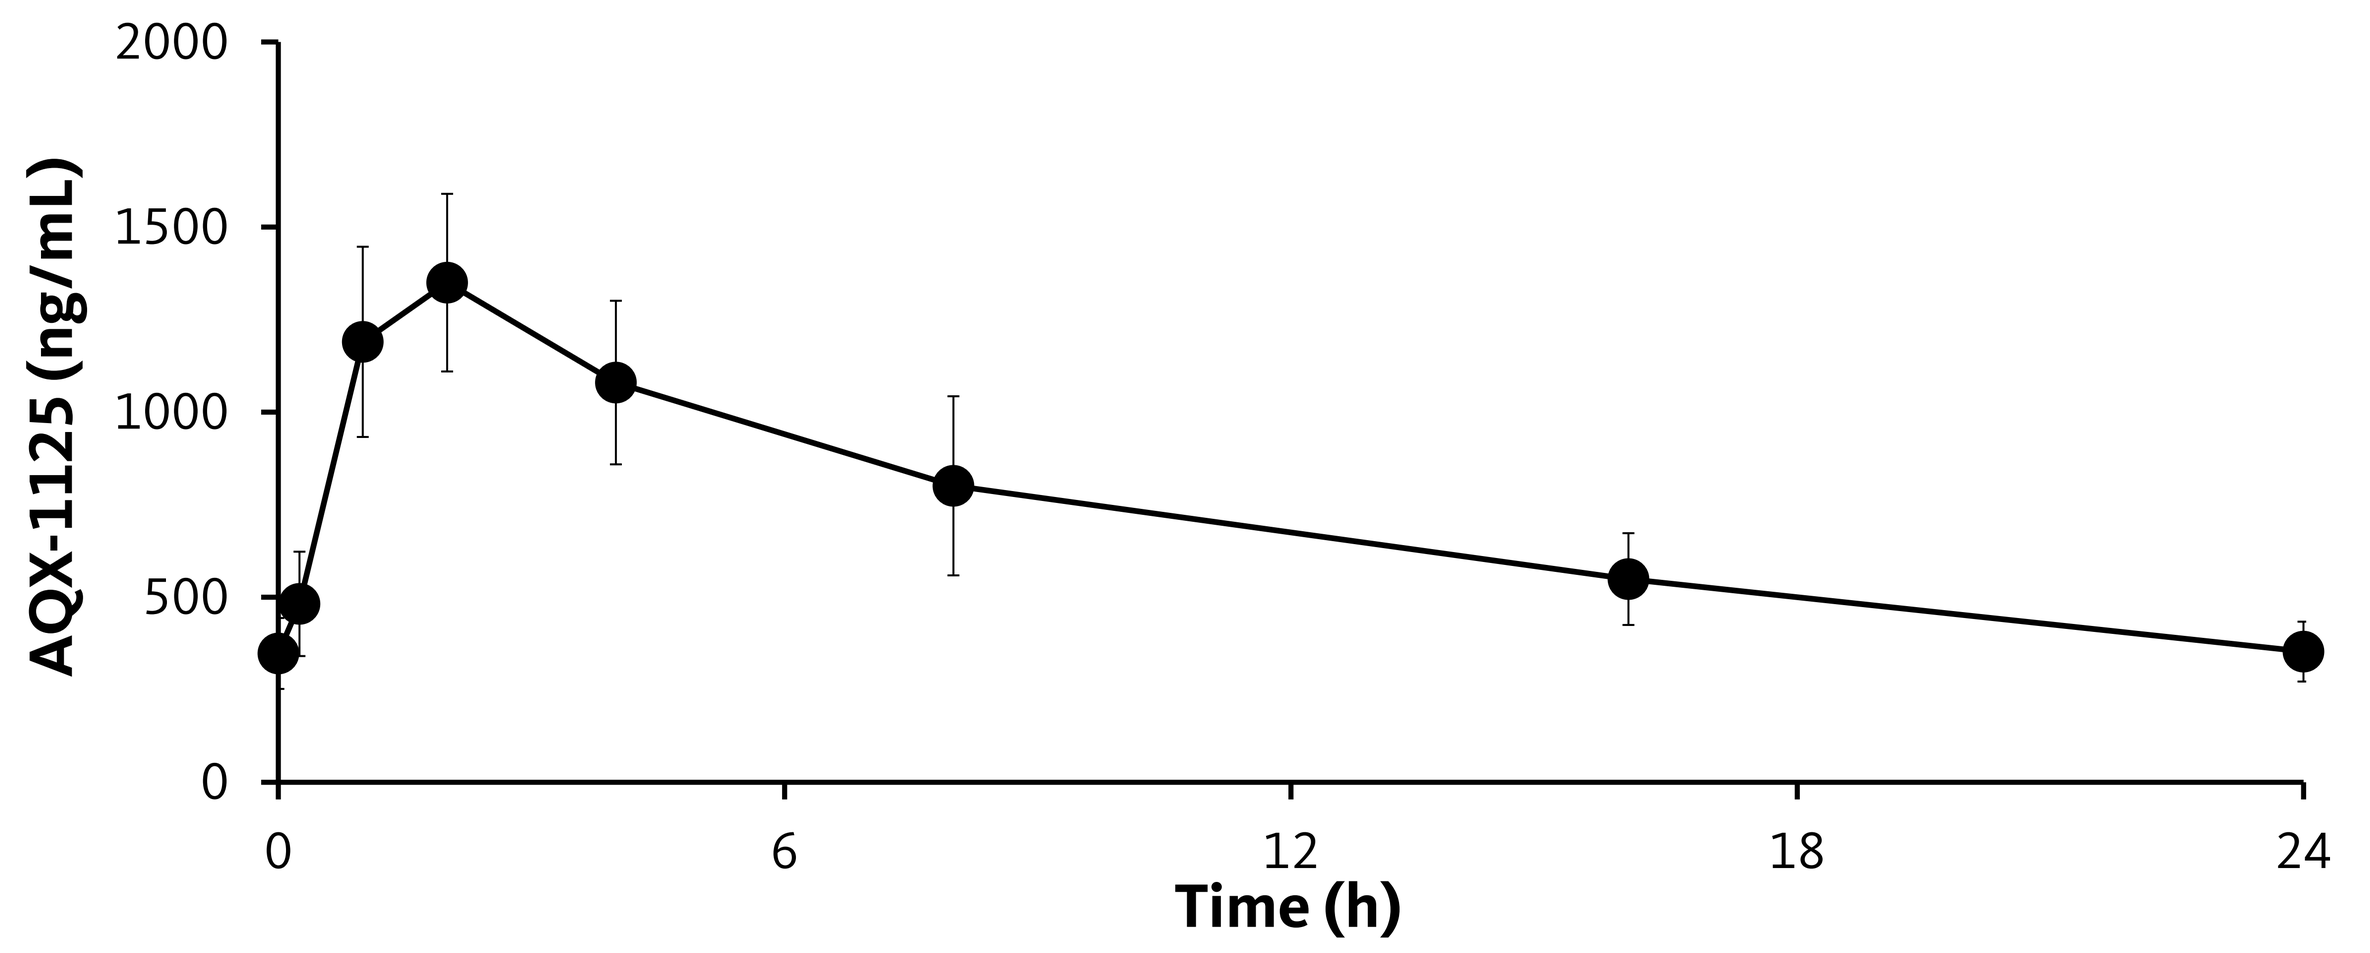
AQX-1125 (ng/mL)
2000
1500
1000
500
0
0
6
12
18
24
Time (h)

Figure 5. Pharmacokinetics of AQX-1125 (450 mg) on the day of LPS challenge. Pharmacokinetic data showed that AQX-1125 was rapidly absorbed with mean $C_{max,ss}$ and $AUC_{0-24h,ss}$ values of 1350 ng/mL and 16900 h.ng/mL respectively.

PK supports once-daily oral administration regimen

Safety

System Organ Class	450 mg Active (n=18) Events	450 mg Active (n=18) n (%)	Placebo (n=18) Events	Placebo (n=18) n (%)
Total	75	18 (100%)	56	16 (89%)
Nervous system disorders	20	12 (67%)	17	12 (67%)
General disorders and administration site conditions	19	12 (67%)	17	12 (67%)
Gastrointestinal disorders	12	10 (56%)	4	2 (11%)
Respiratory, thoracic and mediastinal disorders	11	7 (39%)	8	6 (33%)
Musculoskeletal and connective tissue disorders	9	9 (50%)	7	7 (39%)
Vascular disorders	2	2 (11%)	2	2 (11%)
Injury, poisoning and procedural complications	1	1 (6%)	1	1 (6%)

Table 3. Treatment-emergent adverse events (TEAE) by system organ class in >1 subject. The majority of the TEAEs were considered mild with no TEAEs considered severe. The most common TEAEs were nervous system disorders and general disorders and administration site conditions which were reported in 12 subjects on active treatment and in 12 subjects on placebo treatment. Gastrointestinal TEAEs were reported in 10 subjects on active treatment and only 2 subjects on placebo treatment.

Most AEs related to LPS challenge; GI AEs mild, intermittent, short duration and self-resolving

Clinical Development of AQX-1125

Current Phase II: Studies

- Interstitial Cystitis/Bladder Pain Syndrome "LEADERSHIP" Study
 - Parallel group, randomized, double-blind, placebo-controlled, multicenter study
 - 6 weeks dosing
 - Bladder pain and IC/BPS symptom endpoints
- COPD Exacerbation "FLAGSHIP" Study
 - Parallel group, randomized, double-blind, placebo-controlled, multicenter study
 - 12 weeks dosing
 - Exacerbation rates and COPD symptom endpoints

Human and animal pharmacodynamic study results are presented in the posters titled:

"The Effects Of The Novel SHIP1 Activator AQX-1125 On Allergen-Induced Responses In Mild To Moderate Asthma"

and

"Prophylactic or Therapeutic AQX-1125, A Small Molecule SHIP1 Activator, Inhibits Bleomycin-Induced Pulmonary Fibrosis"

Summary

SHIP1 is a novel drug target which controls PI3K-driven cellular migration and activation. SHIP1's preferential expression in hematopoietic cells and low degree of homology with SHIP2 reduces the likelihood of off-target, off-tissue toxicities. AQX-1125, a small molecule allosteric SHIP1 activator, with PK properties suited to once per day dosing, inhibits the PI3K pathway resulting in an inhibition of Akt phosphorylation and reduced chemotaxis. AQX-1125 has high water-solubility, good oral bioavailability, minimal metabolism, and distributes to the lung at high concentrations.

In this study, once daily orally administered AQX-1125 inhibited the LPS-induced pulmonary inflammation as measured by the significant inhibition of sputum neutrophil accumulation following challenge with inhaled LPS. The magnitude of effect on sputum neutrophils was 67% reduction (vs placebo) at the 24 h timepoint and this effect was correlated with a statistically significant reduction (62% vs placebo) in sputum IL-6 concentrations. Together with the safety and pharmacokinetics, this study demonstrates the pharmacodynamic activity needed to support further clinical development of AQX-1125 for the treatment of inflammatory respiratory diseases.

References

1. Helgason *et al*. Targeted disruption of SHIP leads to hemopoietic perturbations, lung pathology, and a shortened life span. *Genes & Dev.* 1998;12:1610-1620.
2. Ong *et al*. Small molecule agonists of SHIP1 inhibit the phosphoinositide 3-kinase pathway in hematopoietic cells. *Blood.* 2007;110:1942-1949.

Activate excellence. **Activate** innovation. **Activate** success.